Safety and efficacy of fermentative hyaluronan in knee osteoarthritis: a retrospective study.

Daniel Uebelhart [1] and Stefan Berz [2] .

[1] Department of Rheumatology and Institute of Physical Medicine, University Hospital Zurich, Switzerland,

[2] Maras AG, CRO, Cham, Switzerland

Introduction

▪ The use of intra-articular (i.a.) hyaluronan (HA) in the treatment of osteoarthritis (OA) is now well accepted and is based on the principle of viscosupplementation. Viscosupplementation restores the normal rheological properties of the synovial fluid and hence its protective, lubricating, shock absorbing and barrier functions resulting in improved joint homeostasis. We set up a retrospective survey to collect tolerability, safety and efficacy data following i.a. injections of Ostenil® (TRB Chemedica AG, Munich, Germany).

Methods

A total of 23 centres in Switzerland, including private rheumatologists and orthopaedists, that were known to use i.a. SH regularly were recruited. An independent clinical research organisation monitored the study. Investigators were provided with case report forms (CRF's) and asked to record all available data on knee OA patients treated with i.a. HA within the previous 15-month period. No selection was to be made regarding tolerability, efficacy and joints treated. As some patient records also contained data on other HA formulations, these were collected for comparison with Ostenil®. Ostenil®, a natural, non-chemically modified HA of fermentative origin, and Synvisc®, a chemically modified, cross-linked HA derivative of avian origin, were the main products used in these centres.

Data Analysis

The comparative tests were performed using rank statistics (Mann-Whitney test). Age, gender and knee OA severity were used as covariates to analyse efficacy variables. Efficacy results were assessed per treatment cycle. A standard treatment cycle for Ostenil® was defined as 1 injection/week for 3 to 5 weeks, while the standard treatment cycle for Synvisc® was defined as 1 injection/week for 3 weeks. The tolerability analysis included all treatment cycles of Ostenil® or Synvisc® and statistics are expressed per treatment cycles. The safety analysis included all patients receiving i.a. injections of Ostenil® or Synvisc®. Statistics are expressed per injection (first injection, subsequent injections).

Table 1 Demographic Data

Parameter		Ostenil® group	Synvisc® group	P value
Age (years, mean)		60.8	64.1	p=0.04-0.06
Gender:	Female	221 (59.9%)	32 (47.8%)	p<0.09
	Male	145 (39.3%)	34 (50.8%)	
	Missing	3 (0.8%)	1 (1.5%)	
Knee OA severity:	Mild	17 (6.2%)	1 (1.5%)	p<0.06
	Moderate	129 (46.7%)	26 (40.0%)	
	Severe	130 (47.1%)	38 (58.5%)	

Figure 1

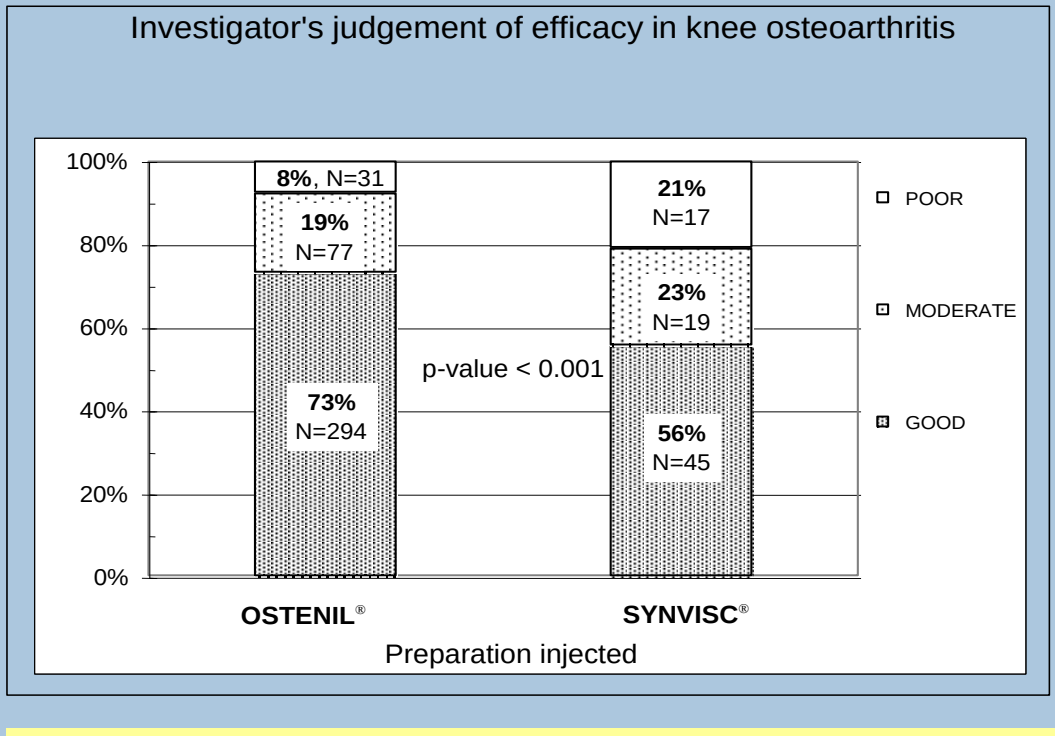
Investigator's judgement of efficacy in knee osteoarthritis
100%
80%
60%
40%
20%
0%
8%, N=31
19%
N=77
73%
N=294
21%
N=17
23%
N=19
56%
N=45
p-value < 0.001
POOR
MODERATE
GOOD
OSTENIL®
SYNVISC®
Preparation injected

Results

Data on 467 patients were obtained of which 436 had symptomatic OA and received one or more i.a. injections of HA into one or both knees. Demographic data are shown in **Table 1**. A total of 2022 i.a. injections were made: 1753 with Ostenil® (86.7%) and 264 with Synvisc® (13.1%). Investigators judged global efficacy as "good" to "moderate" in 92.3% of the Ostenil® treated cases and 79.0% of the Synvisc® treated cases (p<0.001), and "poor" or "insufficient" in 7.7% and 21.0% of the cases, respectively. See **Figure 1**. Efficacy was significantly better (p<0.001) in the Ostenil® group compared to the Synvisc® group. When the comparison is performed for patients having received 3 injections, the efficacy remains significantly better for the Ostenil® group (p=0.03). The investigator's judgement of tolerability was good to moderate in 98.7% of the patients treated with Ostenil® and in 92.6% of the patients treated with Synvisc® See **Table 2**. The incidence of adverse device events (ADE's) in the Synvisc® treated cases was 7.7% compared to 2.1% in the Ostenil® group (p<0.0001) while the incidence of adverse device reactions (ADR's) was 5.1% in the Synvisc® group and 0.7% in the Ostenil® group (p<0.0001). See **Figure 2**. The overall incidence of ADR's with the HA products was 6.1%, with 3.9% in the Ostenil® group and 15.2% in the Synvisc® group. ADR's were significantly more frequent and more severe with Synvisc®.

Figure 2

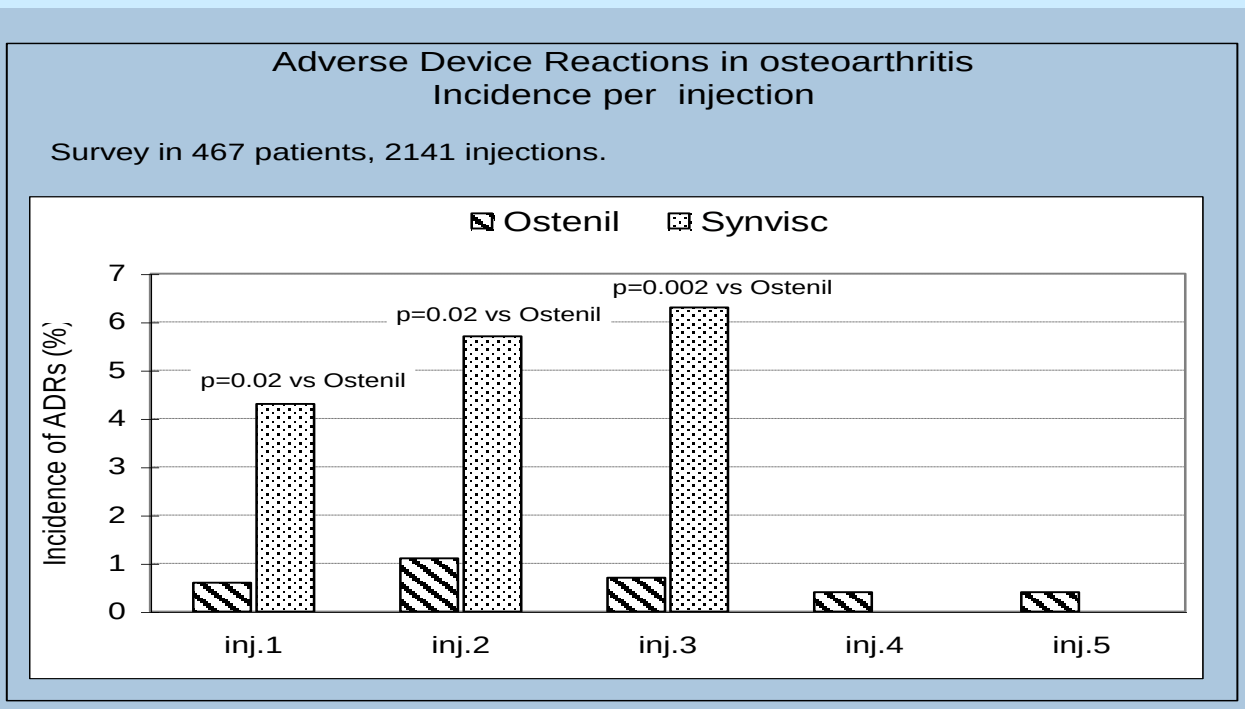
Adverse Device Reactions in osteoarthritis
Incidence per injection
Survey in 467 patients, 2141 injections.
Ostenil
Synvisc
Incidence of ADRs (%)
7
6
5
4
3
2
1
0
p=0.02 vs Ostenil
p=0.02 vs Ostenil
p=0.002 vs Ostenil
inj.1
inj.2
inj.3
inj.4
inj.5

Table 2 Assessment of tolerability in the treated OA knee by preparation

	Good		Moderate		Poor		Total	
	N	%	N	%	N	%	N	%
Ostenil® group	289	98.0	2	0.7	4	1.4	295	100.0
Synvisc® group	61	89.7	2	2.9	5	7.4	68	100.0

Conclusion: The results of this retrospective study indicate that Ostenil®, which contains a natural, non-chemically modified HA of fermentative origin, is a safe and effective therapy for knee OA, and support previously published data[1] indicating that i.a. injection of chemically modified cross-linked HA derivative of avian origin (Synvisc®) is associated with a higher incidence of adverse device reactions.

Reference: Maheu E, Ayral X, Dougados M. A hyaluronan preparation (500-730 kDa) in the treatment of osteoarthritis: a review of clinical trials with Hyalgan. Int J Clin Pract 2002; 56(10):804-13.

Acknowledgement: The authors would like to thank Leanne Pobjoy for here help in preparing the poster.