Long-term outcome of synovial fluid substitution with Viscoseal® post-arthroscopy

Prof. Dr. med. Harald Hempfling
Berufsgenossenschaftliche Unfallklinik, D-82418 Murnau am Staffelsee

Aim

To investigate the efficacy and safety of post-arthroscopic intra-articular sodium hyaluronate in patients treated with lavage / debridement because of persisting severe pain in the knee joint.

Rationale

Arthroscopic lavage is a therapeutic option when conservative treatment fails. The procedure flushes detritus, inflammatory cells and molecule out from the joint and allows debridement of the cartilage where necessary.

The flushing solutions used in lavage may exert negative effects on the metabolism and structure of the joint cartilage, because Hyaluronic acid (HA), an important constituent of synovial fluid and joint cartilage, is removed. A temporary substitute of the synovial fluid, based on HA (Viscoseal®) has been developed in order to protect the tissues of the joint and to promote recovery of joint function post arthroscopy.

Patients, Methods and Material

Prospective, randomised, masked-observer study:
Lavage (A) vs. Lavage + Hyaluronic Acid (A+HA)

Patients:
- 80 patients with severe pain in the knee joint, persisting for at least 6 months (VAS pain > 50 mm)
- mean age : 60.9 ± 8.1 years

Study product and administration:
- 50 mg/10ml Na-hyaluronate obtained by fermetation (Viscoseal). HA-concentration similar to the one in synovial fluid
- One intra-articular administration, immediately after arthroscopy

Safety:
- Documentation of evident clinical reactions

	pre (V0)	intra-OP (V1)	6h post (V2)	2 weeks (V3)	3 months (V4)	1 year (V5)
Clinical findings, In- and exclusion criteria	X					
Outerbridge-findings		X				
VAS (investigator)	X		X	X		
VAS (pain diary patient)			daily for 2 weeks			
Clinical Global Impression (CGI)			X	X	X	X
Problems when walking 100 m	X		X	X	X	X
Pain when walking 100 m	X		X	X	X	X
Pain at night	X		X	X	X	X
Safety			X	X	X	X

Results

- No unexpected or adverse events
- No influence of Outerbridge-stadium and debridement on patient outcome
- Marked reduction of pain under both treatments (A-group and A+HA-group) (Fig. 2-4)
- No intergroup differences in the acute p.a. phase (V0-V3). (Fig 1-4)
- Three months after treatment (V4) relapse in the A-group (Mann-Whitney-statistics: shown superiority of the A+HA-group) (Fig. 1-4)
- Clear intergroup differences in the long term course (V0-V5). One year after treatment (V5) all efficacy parameters in favour of the A+HA-group (Mann-Whitney-statistics: proven superiority of the A+HS-group) :
 - Clinical Global Impression (Fig. 1a and b)
 - Problems, when walking 100 m (Fig. 2)
 - Pain, when walking 100 m (Fig. 3)
 - Pain at night (Fig. 4)

Fig.1a: Clinical global impression (patient) Mann-Whitney-statistics

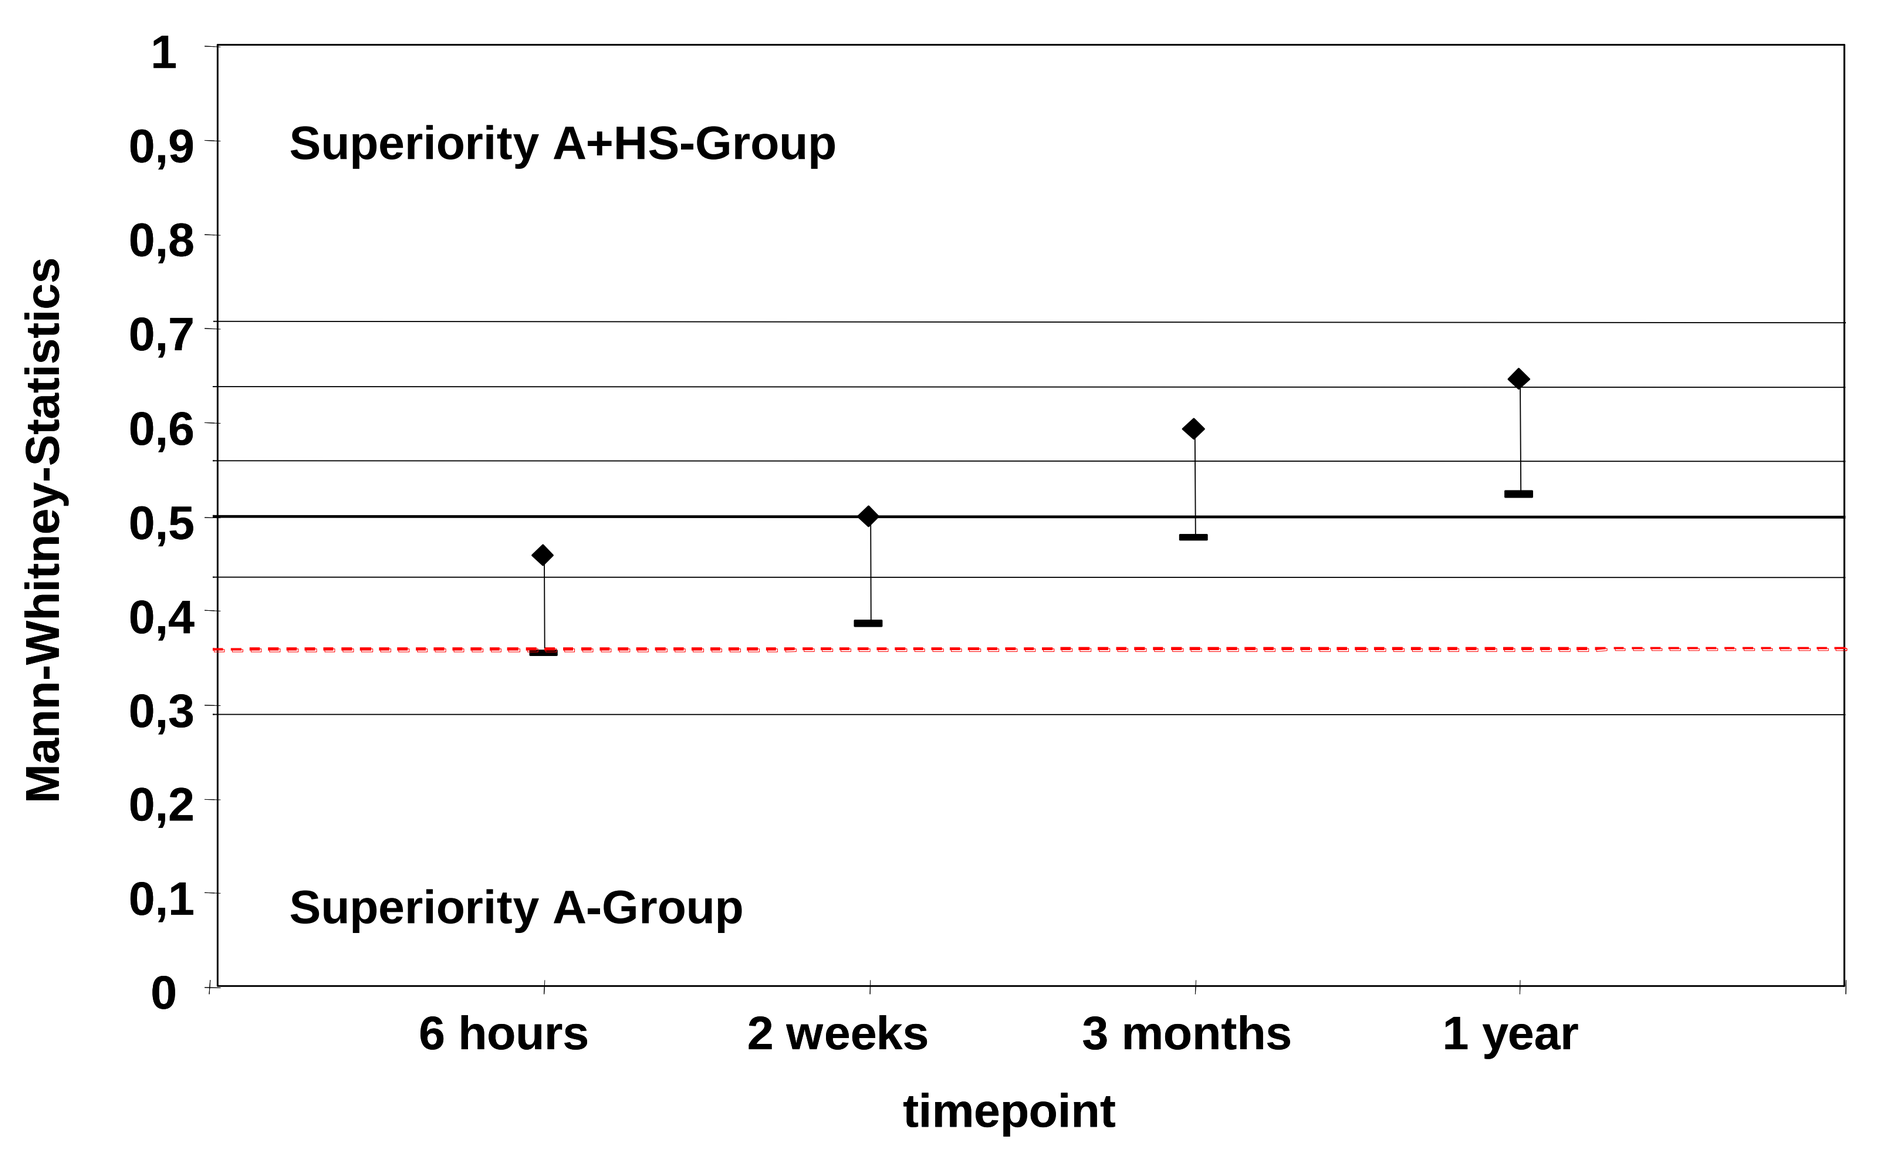
Superiority A+HS-Group
Superiority A-Group
Mann-Whitney-Statistics
1
0,9
0,8
0,7
0,6
0,5
0,4
0,3
0,2
0,1
0
6 hours
2 weeks
3 months
1 year
timepoint

Figure 1b: Clinical global impression (patient)

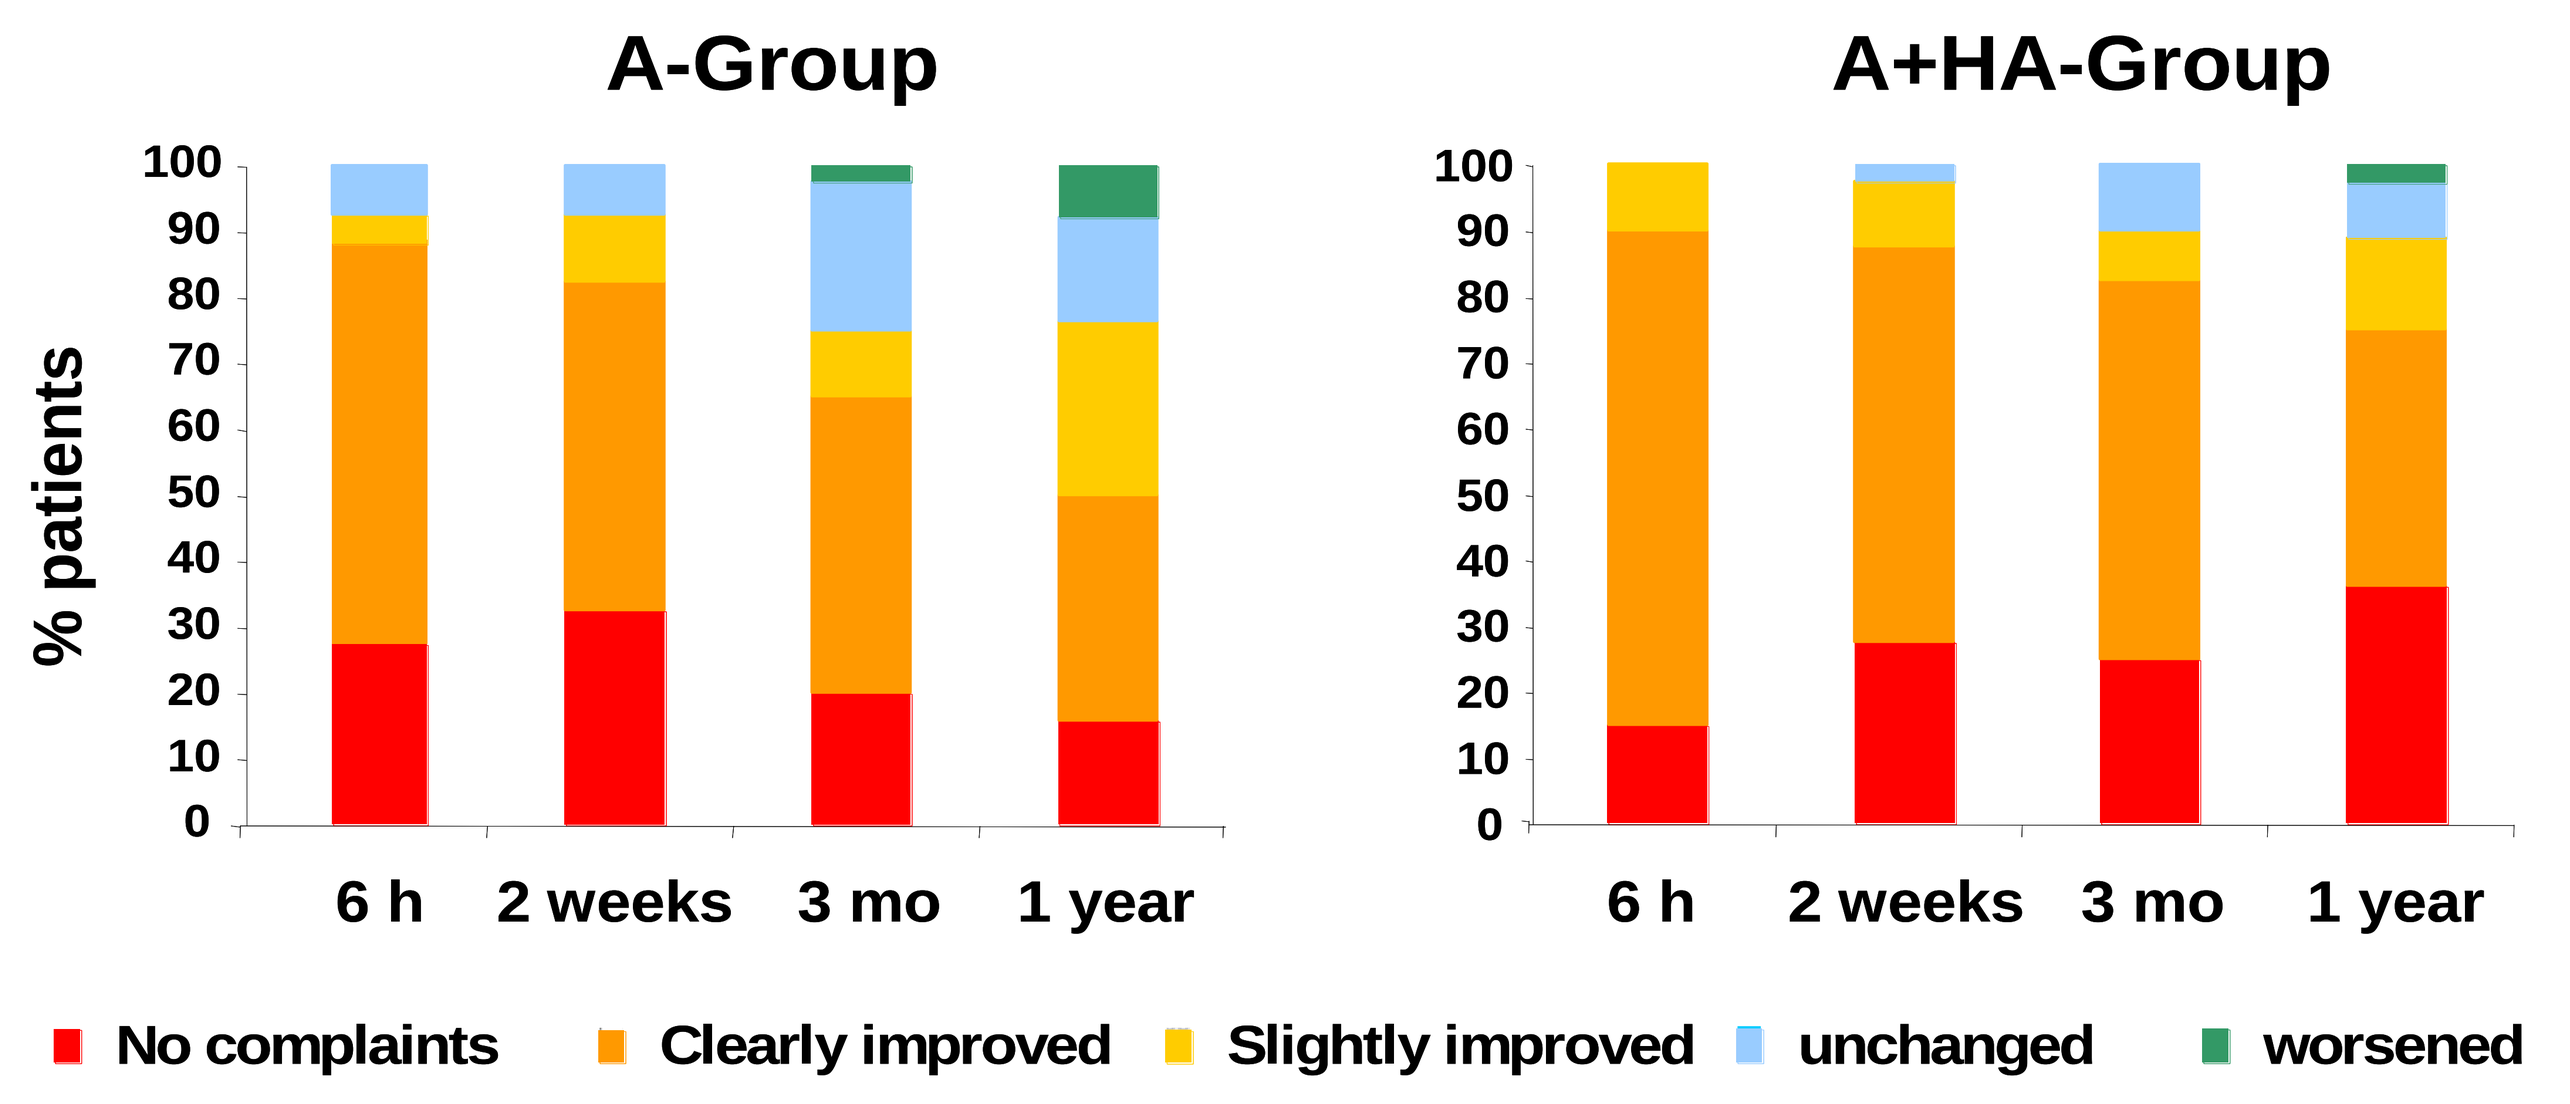
A-Group
A+HA-Group
% patients
6 h
2 weeks
3 mo
1 year
No complaints
Clearly improved
Slightly improved
unchanged
worsened

Figure 2: Problems, when walking 100 m - one year after treatment

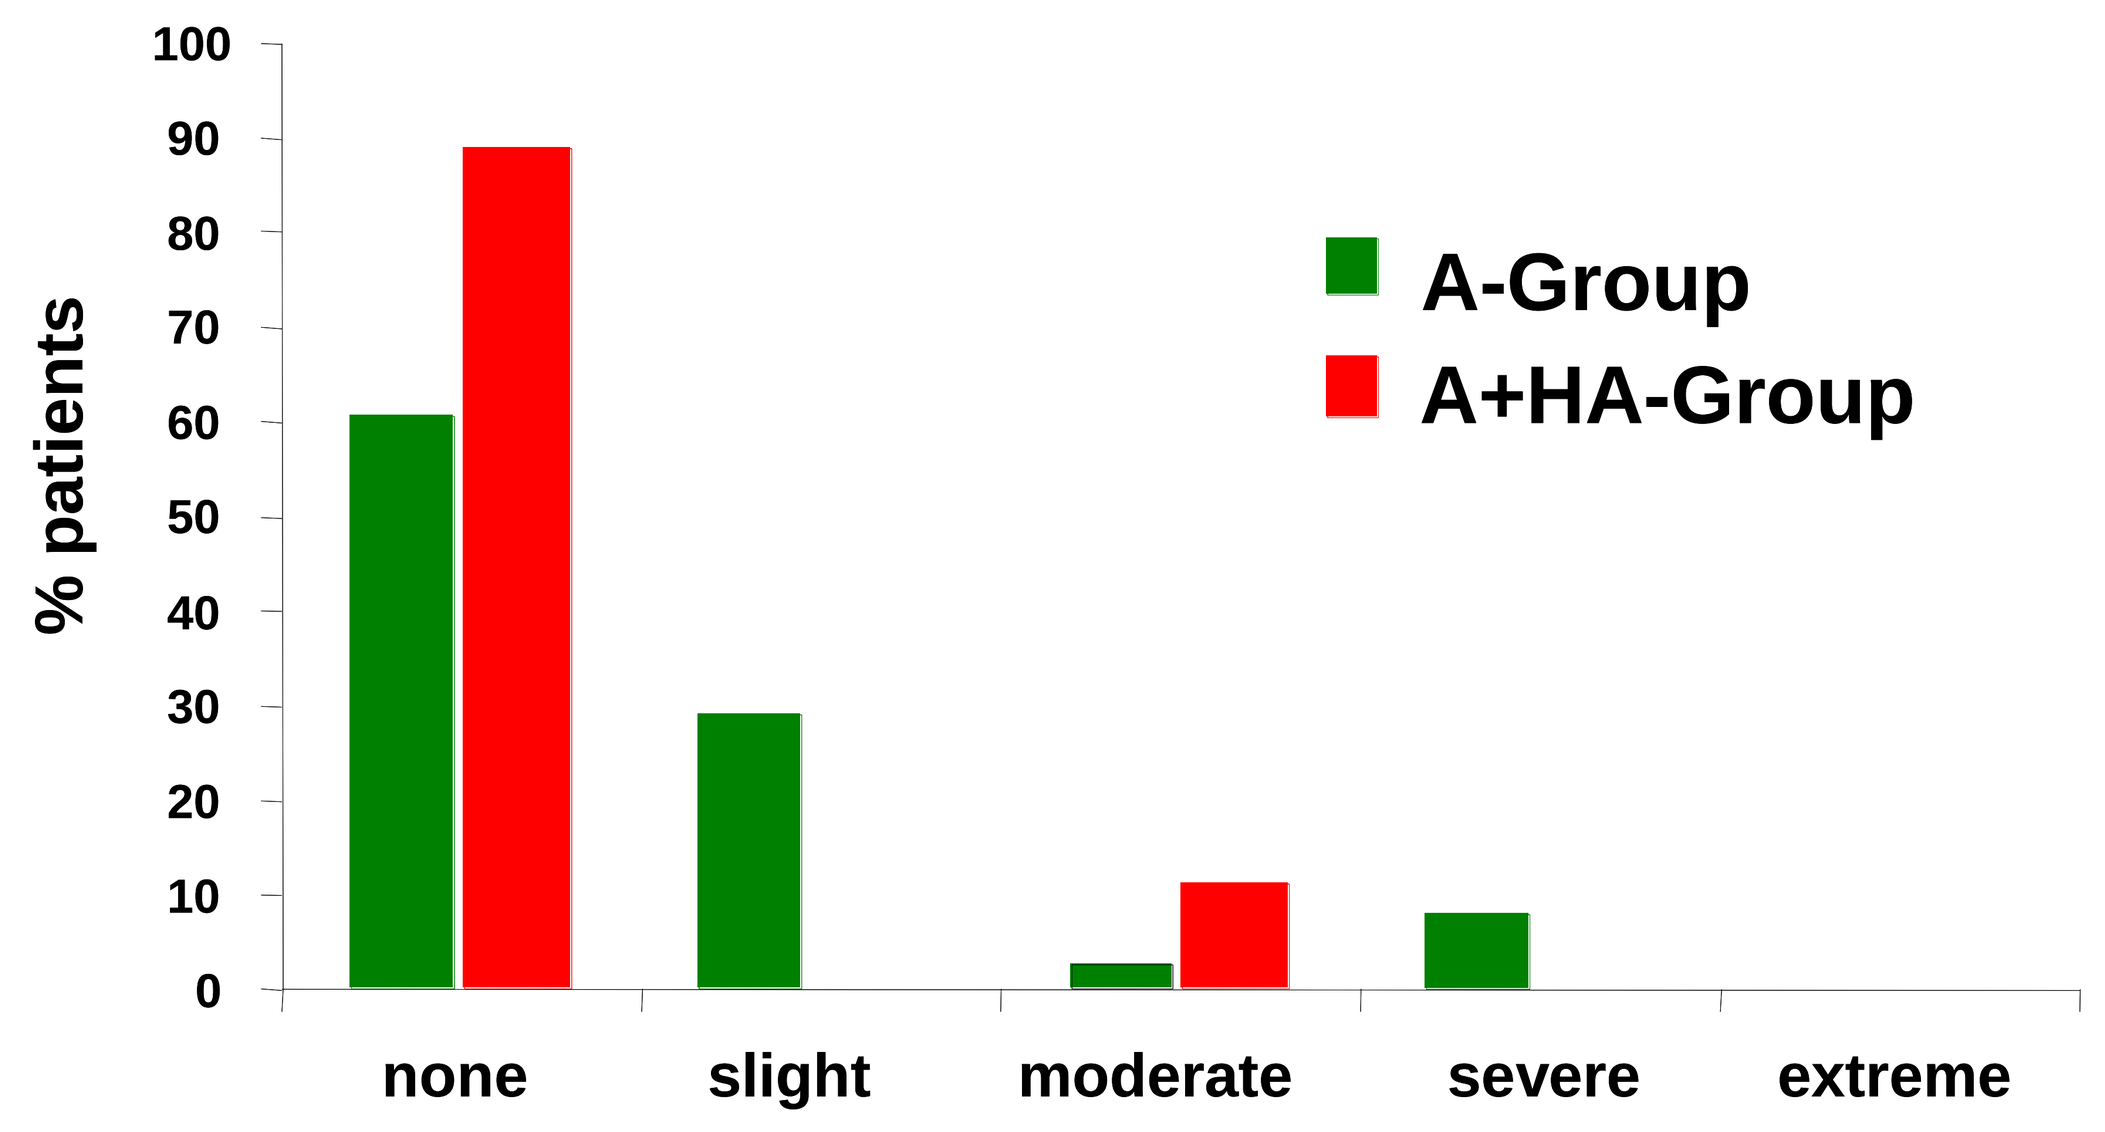
% patients
A-Group
A+HA-Group
none
slight
moderate
severe
extreme

Figure 3: Pain, when walking 100 m - throughout study period

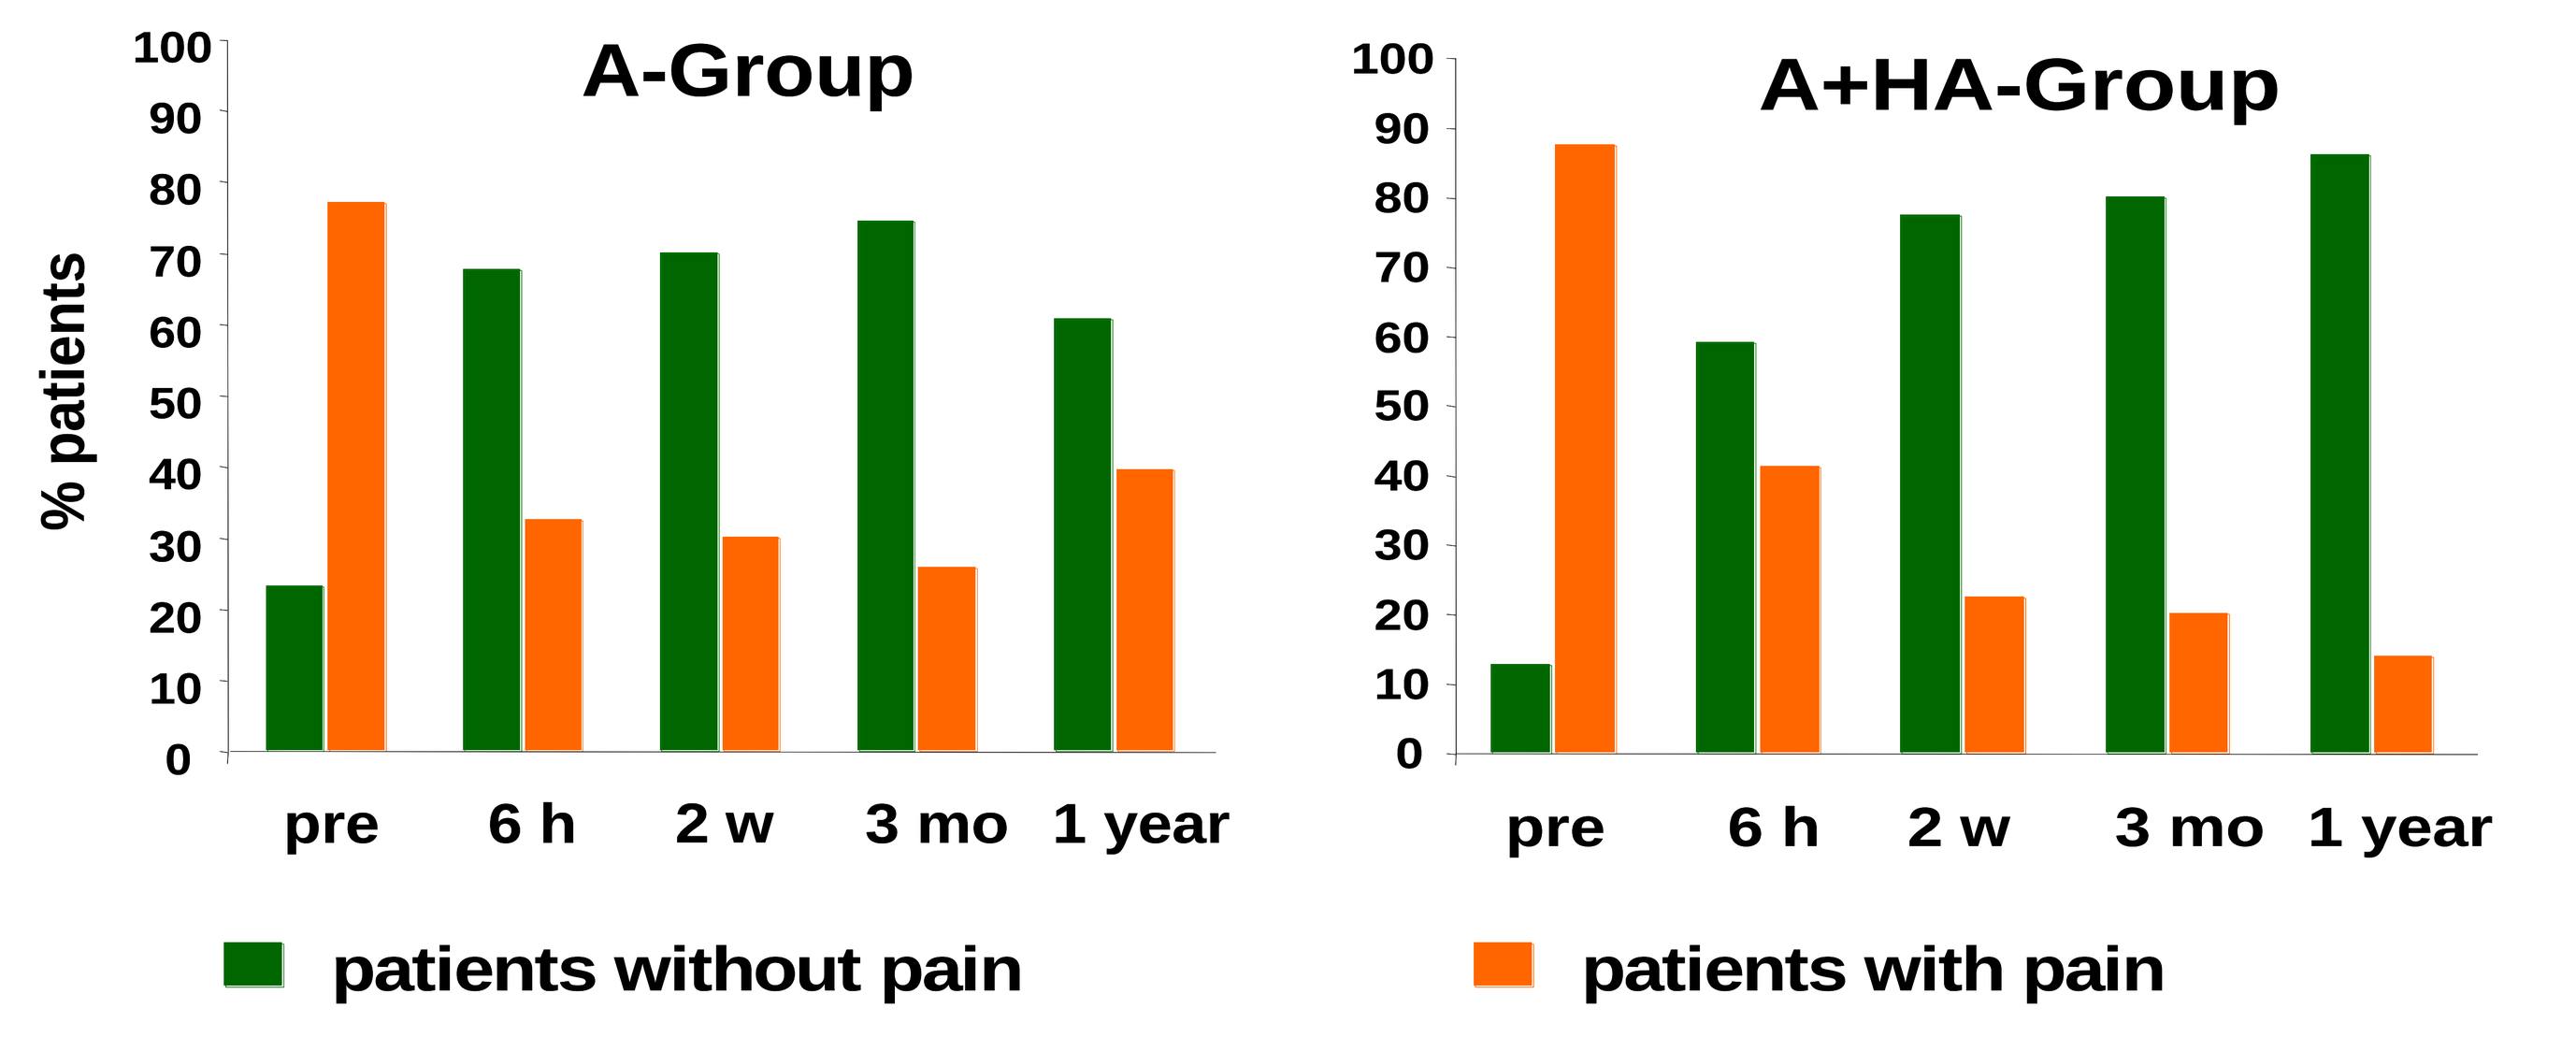
A-Group
A+HA-Group
% patients
pre
6 h
2 w
3 mo
1 year
patients without pain
patients with pain

Figure 4: Pain at night - throughout study period

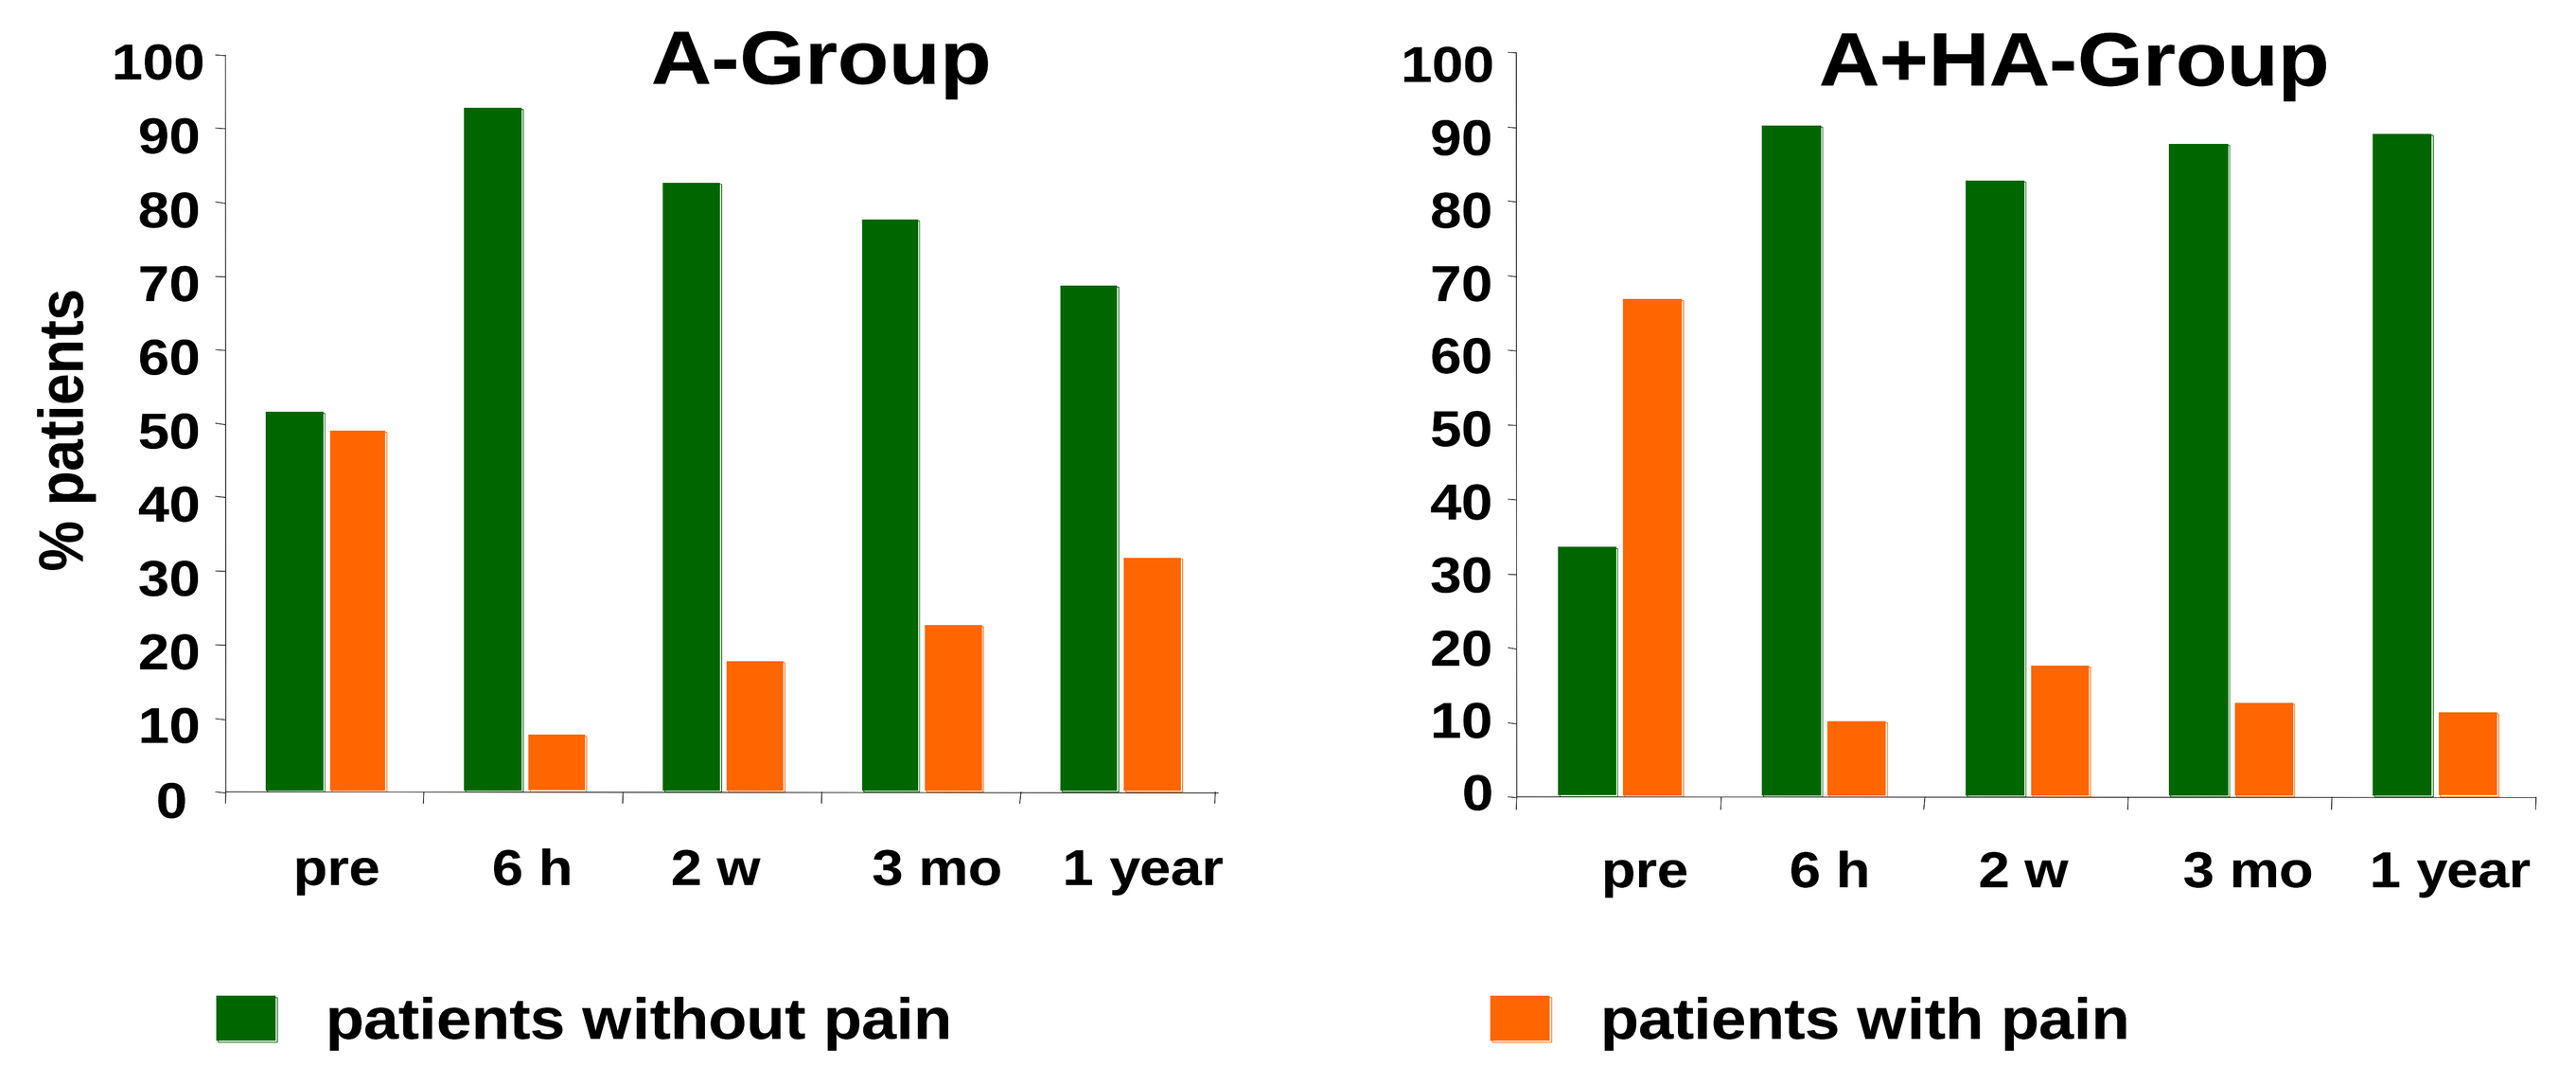
A-Group
A+HA-Group
% patients
pre
6 h
2 w
3 mo
1 year
patients without pain
patients with pain

Summary

- Arthroscopic lavage of the knee joint, in combination with debridement where necessary, reduces chronic pain and restricted mobility for a long period of time.
- Intra-articular Hyaluronic acid (Viscoseal®) “stabilises” the improvement achieved through lavage when administered as a temporary synovia substitute immediately post arthroscopy.
- A Risk-Benefit-Evaluation is in favour of i.a. Hyaluronic acid (Viscoseal®) post arthroscopy.
- Further studies are recommended for understanding better the mechanisms of the carry-over effect of intra-articular Hyaluronic acid.

References

- VAD VB, Bhat AL, Sculo TP, Wickiewicz TL. Management of knee osteoarthritis: knee lavage combined with hylan versus hylan alone. *Arch Phys Med Rehabil* 84:634-637, 2003
- MATTHIES B. Postoperative Hyaluronsäure – Akutsubstitution optimiert Therapieerfolg. *Sport Orthop Traumatol* 18:190-191, 2002